Pharmacokinetic Profile of a Novel Low Dose Extended Release Formulation of AGB101 (Levetiracetam)

Sharon Rosenzweig-Lipson, PhD[1], Steven Mulcahy[1], Ken Payie, PhD[1], Sarah Bullock[2], Elsie Melsopp, PhD[3], Jack James[3], Michela Gallagher, PhD[1]
AgeneBio, Inc[1], Baltimore, MD, USA Worldwide Clinical Trials[2], Early Phase Services, San Antonio, TX, USA AAI Pharma[3], Wilmington, NC, USA

Background

A distinctive condition in the aMCI patient population is neural overactivity localized to the hippocampal formation by functional magnetic resonance imaging (fMRI) (Dickerson et al, 2005; Yassa et al., 2010). In clinical studies, the magnitude of such hippocampal overactivity longitudinally predicts subsequent cognitive decline/conversion to a dementia diagnosis (Miller et al., 2008; Putcha et al., 2011; Huijbers et al, 2015).

In a Phase II study (Bakker et al., 2012, 2015), levetiracetam (125 – 500 mg total daily dose) was evaluated in patients with aMCI to determine its effects on hippocampal overactivity and on memory function in a behavioral pattern separation task performed during fMRI scans. Low doses of levetiracetam (125 – 250 mg daily), but not the high dose (500 mg daily) reduced hippocampal overactivity and significantly improved task related memory performance. Drug levels in aMCI patients were determined to be 2.9 ± 0.29 ug/mL (mean ± sem) for the 125 mg cohort and 4.4 ± 0.53 ug/mL for the 250 mg cohort. The ineffective dose of 500 mg provided a drug level of 7.91 ± 0.92 ug/mL. These levels of drug exposure are well below typical ranges for efficacy of levetiracetam as an antiepileptic agent, where doses of 1000-3000 mg/day are typical, achieving levels of 10-40 mg/mL. The efficacious exposures observed in aMCI patients were consistent with animal studies in age-impaired rats in which exposures of 1.9 – 3.9 mg/mL were efficacious and an exposure of 7.8 mg/mL was ineffective.

We conducted a crossover food effect study of two novel low dose extended release formulations of AGB101 (low dose levetiracetam) at doses of 190 and 220 mg. The purposes of the study were to confirm the exposures associated with efficacy in the Phase II trial and in age-impaired rats were observed from the novel formulations and were maintained over the course of the daytime hours, to confirm the formulation PK profile was consistent with once daily dosing and to determine if there were any food effects on the PK profile.

Methods

This was an open-label, randomized, two-group, single-dose, two-period crossover, food-effect study in which 56 healthy adult subjects received a single dose of AGB101 extended-release tablet (either 190 or 220 mg) under fed conditions in one study period and a separate single dose of AGB101 extended-release tablet under fasted conditions in another study period.

Fed treatment: Following an overnight fast of at least 10 hours, subjects consumed a Food and Drug Administration (FDA) standard high-calorie, high-fat breakfast meal 30 minutes prior to administration of the study drug.

Fasted treatment: Subjects were dosed after an overnight fast of at least 10 hours.

Each drug administration was separated by a washout period of at least 7 days.

Blood samples (1 × 6 mL) were collected in Vacutainer tubes containing K_2-EDTA as a preservative, at 0 (predose) and at 1.0, 2.0, 3.0, 4.0, 4.5, 5.0, 5.5, 6.0, 6.5, 7.0, 8.0, 9.0, 10, 12, 18, 24, 32, and 48 hours post dosing.

Low, but not high dose, AGB101 attenuates hippocampal overactivity and improves memory in a pattern separation task (Bakker et al 2012, 2015)

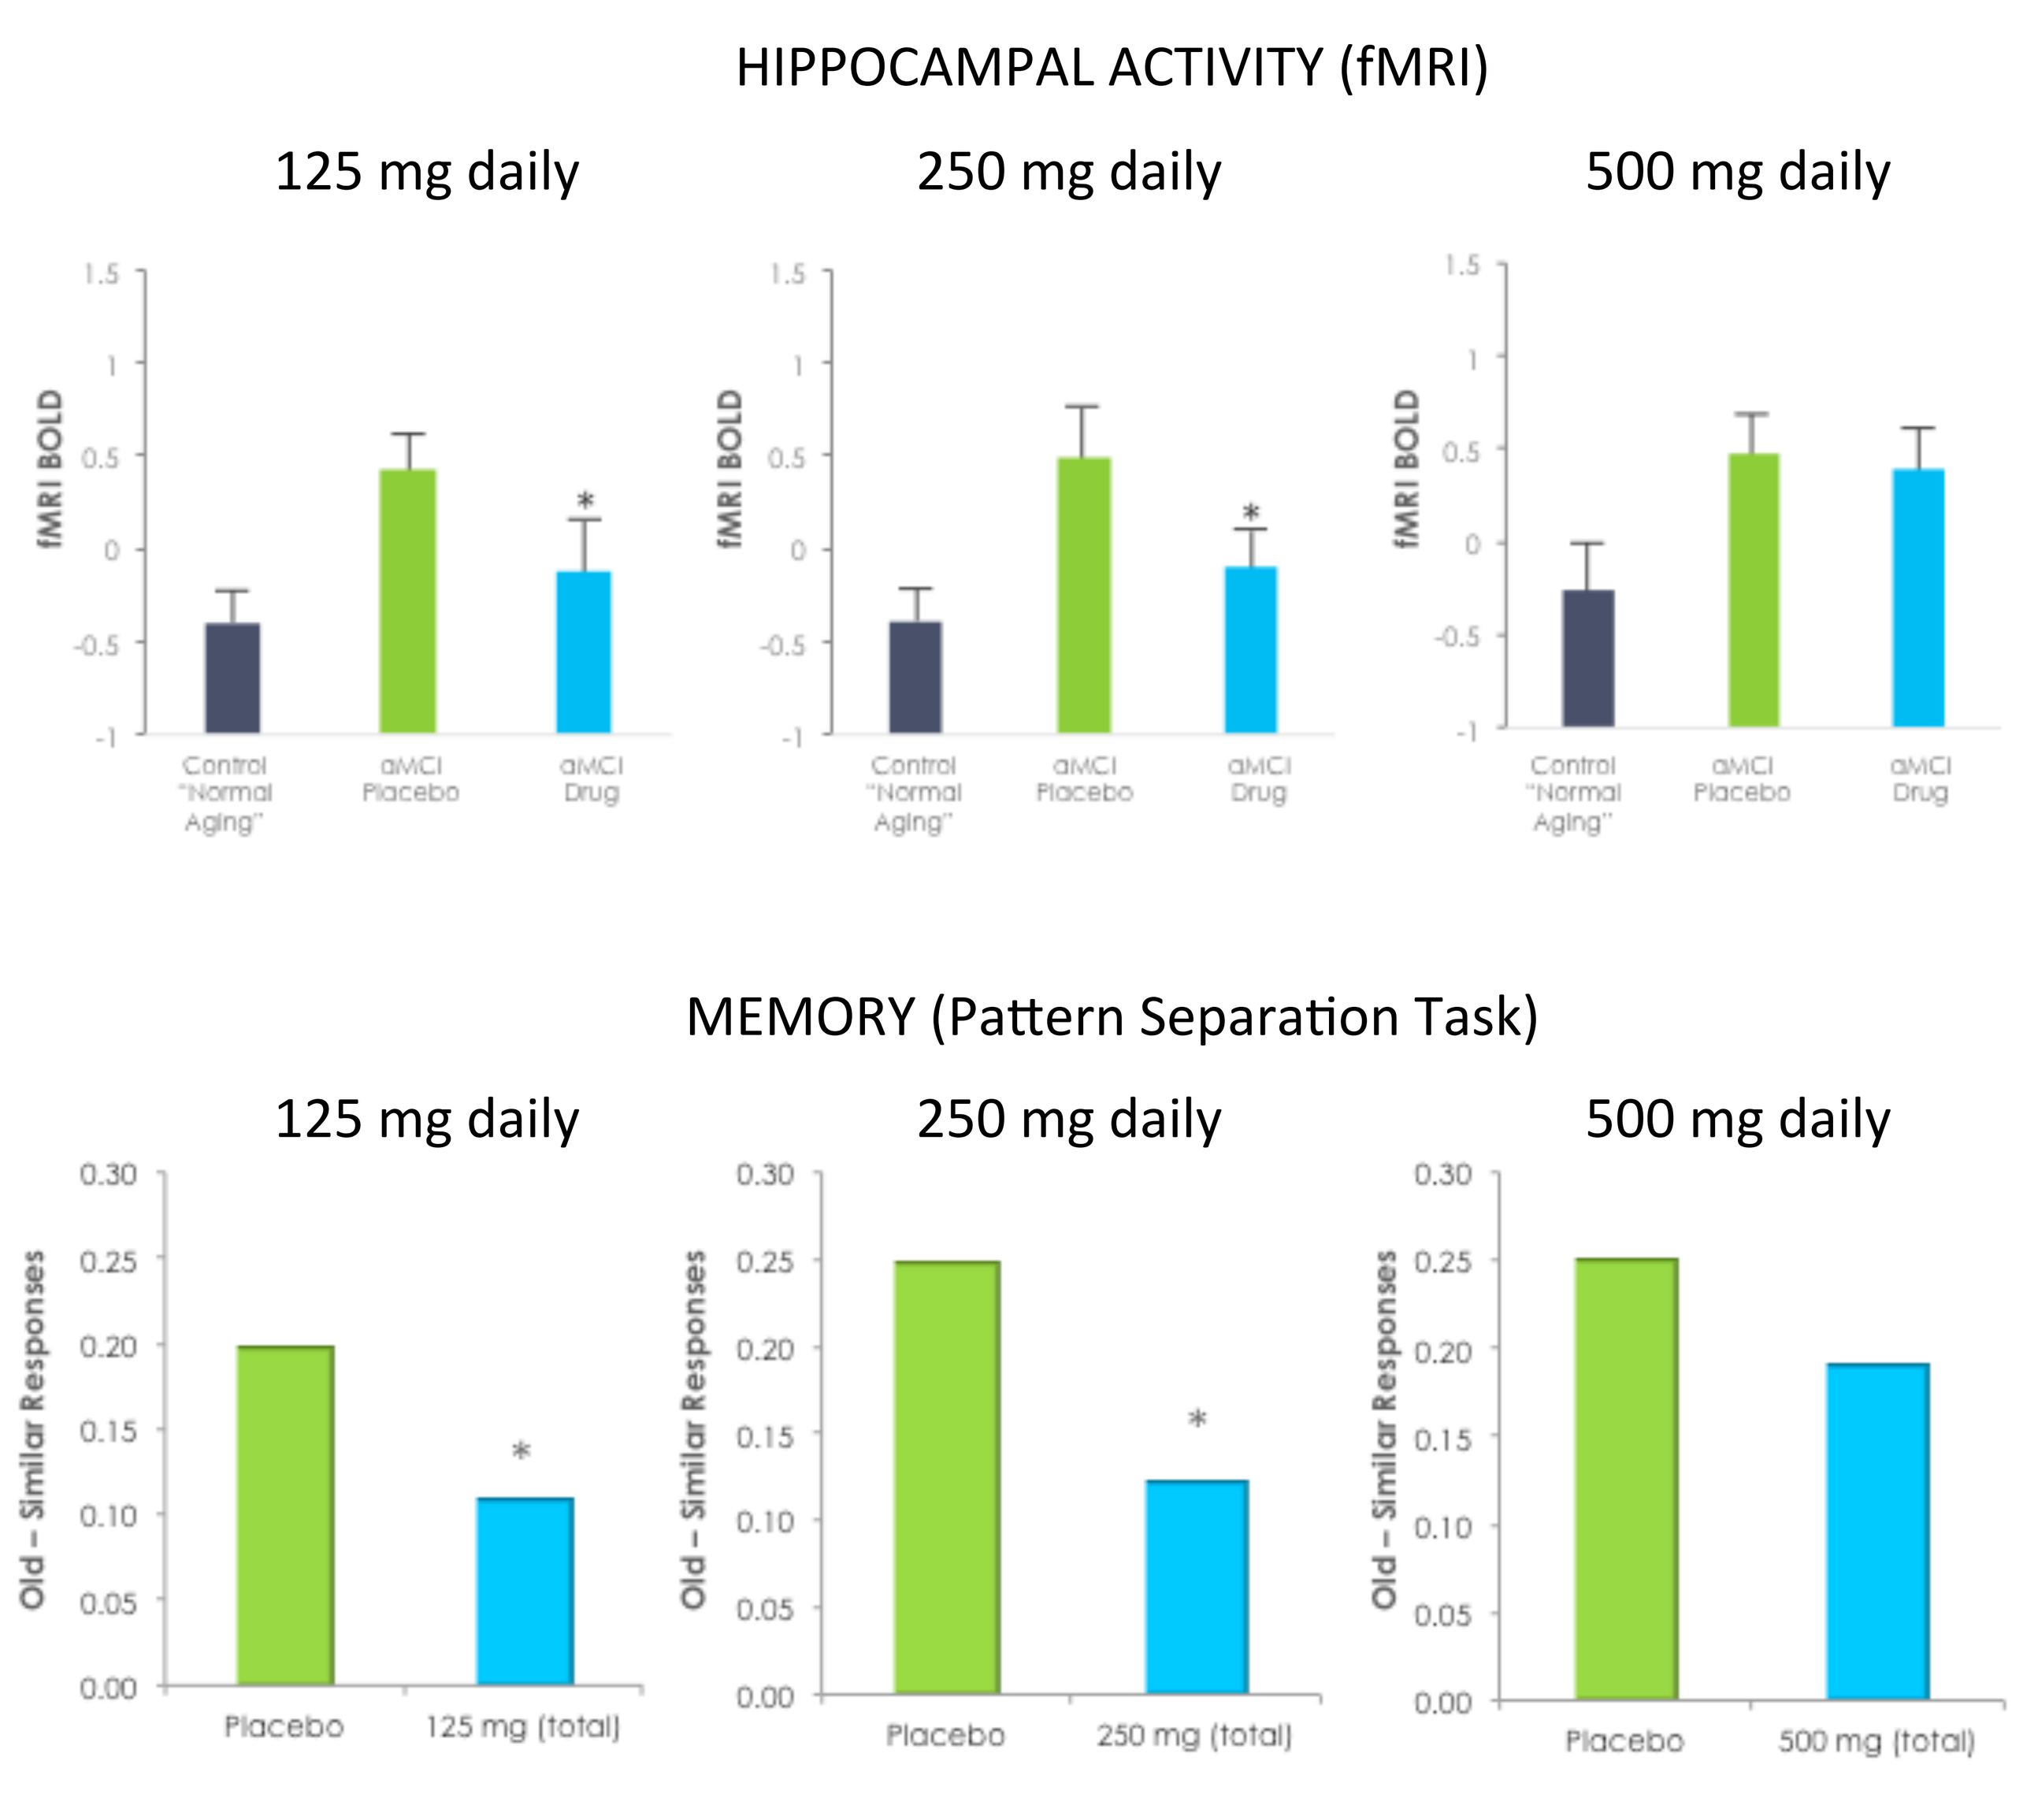
HIPPOCAMPAL ACTIVITY (fMRI)
125 mg daily
250 mg daily
500 mg daily
fMRI BOLD
Control "Normal Aging"
aMCI Placebo
aMCI Drug
MEMORY (Pattern Separation Task)
125 mg daily
250 mg daily
500 mg daily
Old – Similar Responses
Placebo
125 mg (total)
250 mg (total)
500 mg (total)

- Low doses (125-250 mg daily) attenuate hippocampal overactivity and improve pattern separation.
- A high dose (500 mg daily) fails to improve either measure.

AGENEBIO

Study Goals

- Confirm exposures consistent with Phase 2 study exposures
- Confirm formulation PK consistent with once daily dosing
- Determine any food effects on PK

Efficacious exposures similar in Age-Impaired Rats and aMCI patients. Ineffective exposures also similar.

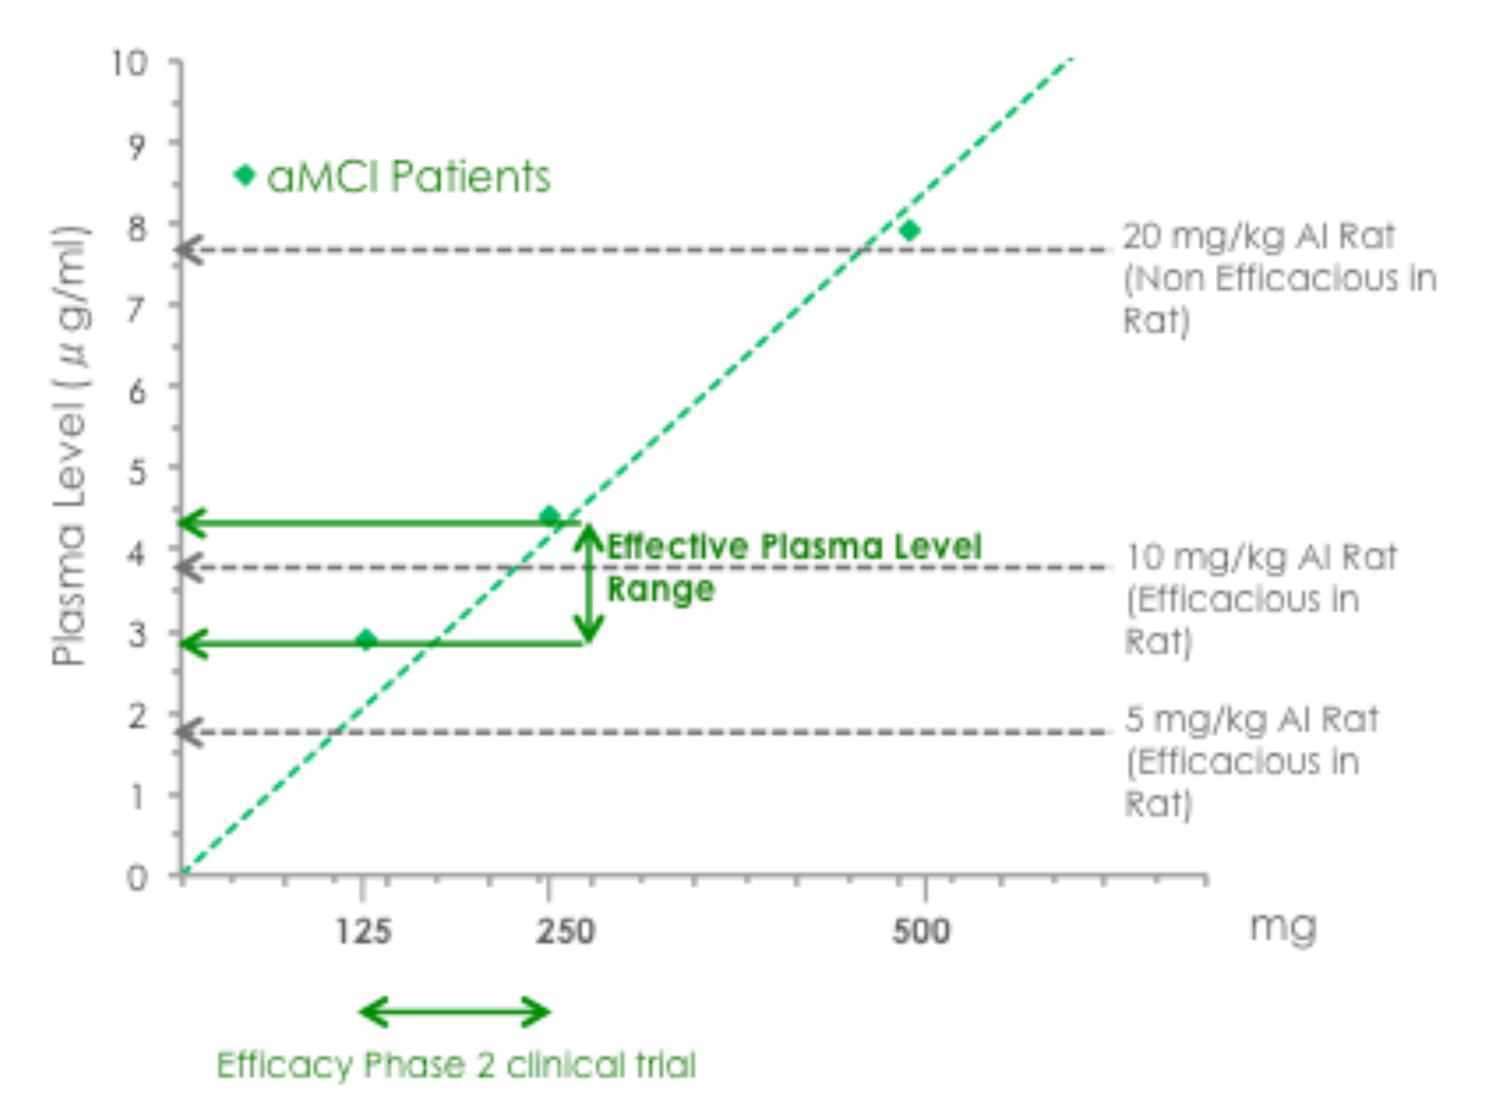
aMCI Patients
Plasma Level (µg/ml)
20 mg/kg AI Rat (Non Efficacious in Rat)
Effective Plasma Level Range
10 mg/kg AI Rat (Efficacious in Rat)
5 mg/kg AI Rat (Efficacious in Rat)
125
250
500
mg
Efficacy Phase 2 clinical trial

	Observed Efficacy (µg/mL)
Age-Impaired Rats	1.9 – 3.9
aMCI	2.9 – 4.4

	No Efficacy (µg/mL)
Age-Impaired Rats	7.8
aMCI	7.9

Steady State Formulation Goals:

- Preferred range based on aMCI between 2.9 and 4.4 µg/mL
- Acceptable range based AI rats and aMCI between 1.9 and 4.4 µg/mL

190 mg and 220 mg Extended Release AGB101 Single Dose PK profile (Fasted A1 vs Fed B1)

190 mg

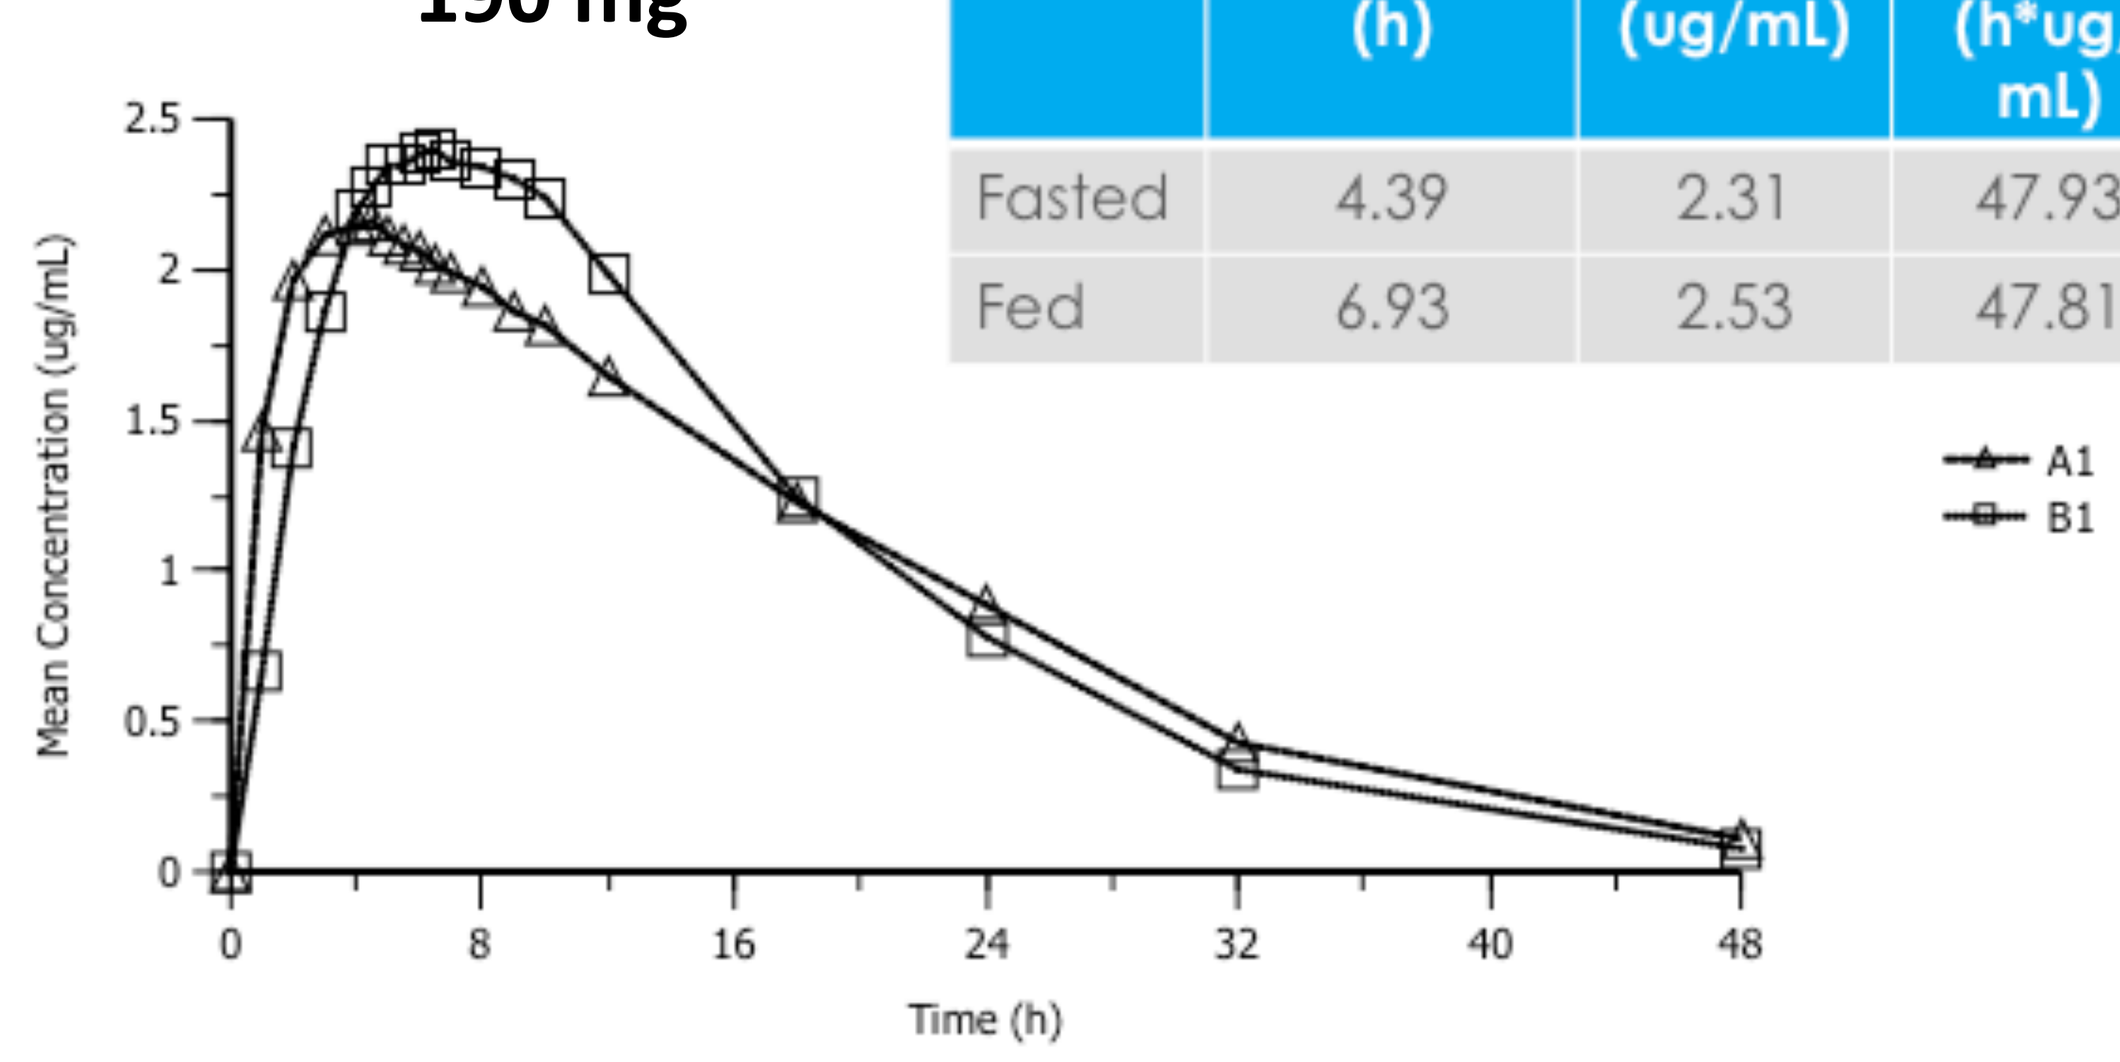
Mean Concentration (ug/mL)
Time (h)
A1
B1

	Tmax (h)	Cmax (ug/mL)	AUCinf (h*ug/mL)	T1/2 (h)
Fasted	4.39	2.31	47.93	8.09
Fed	6.93	2.53	47.81	7.70

220 mg

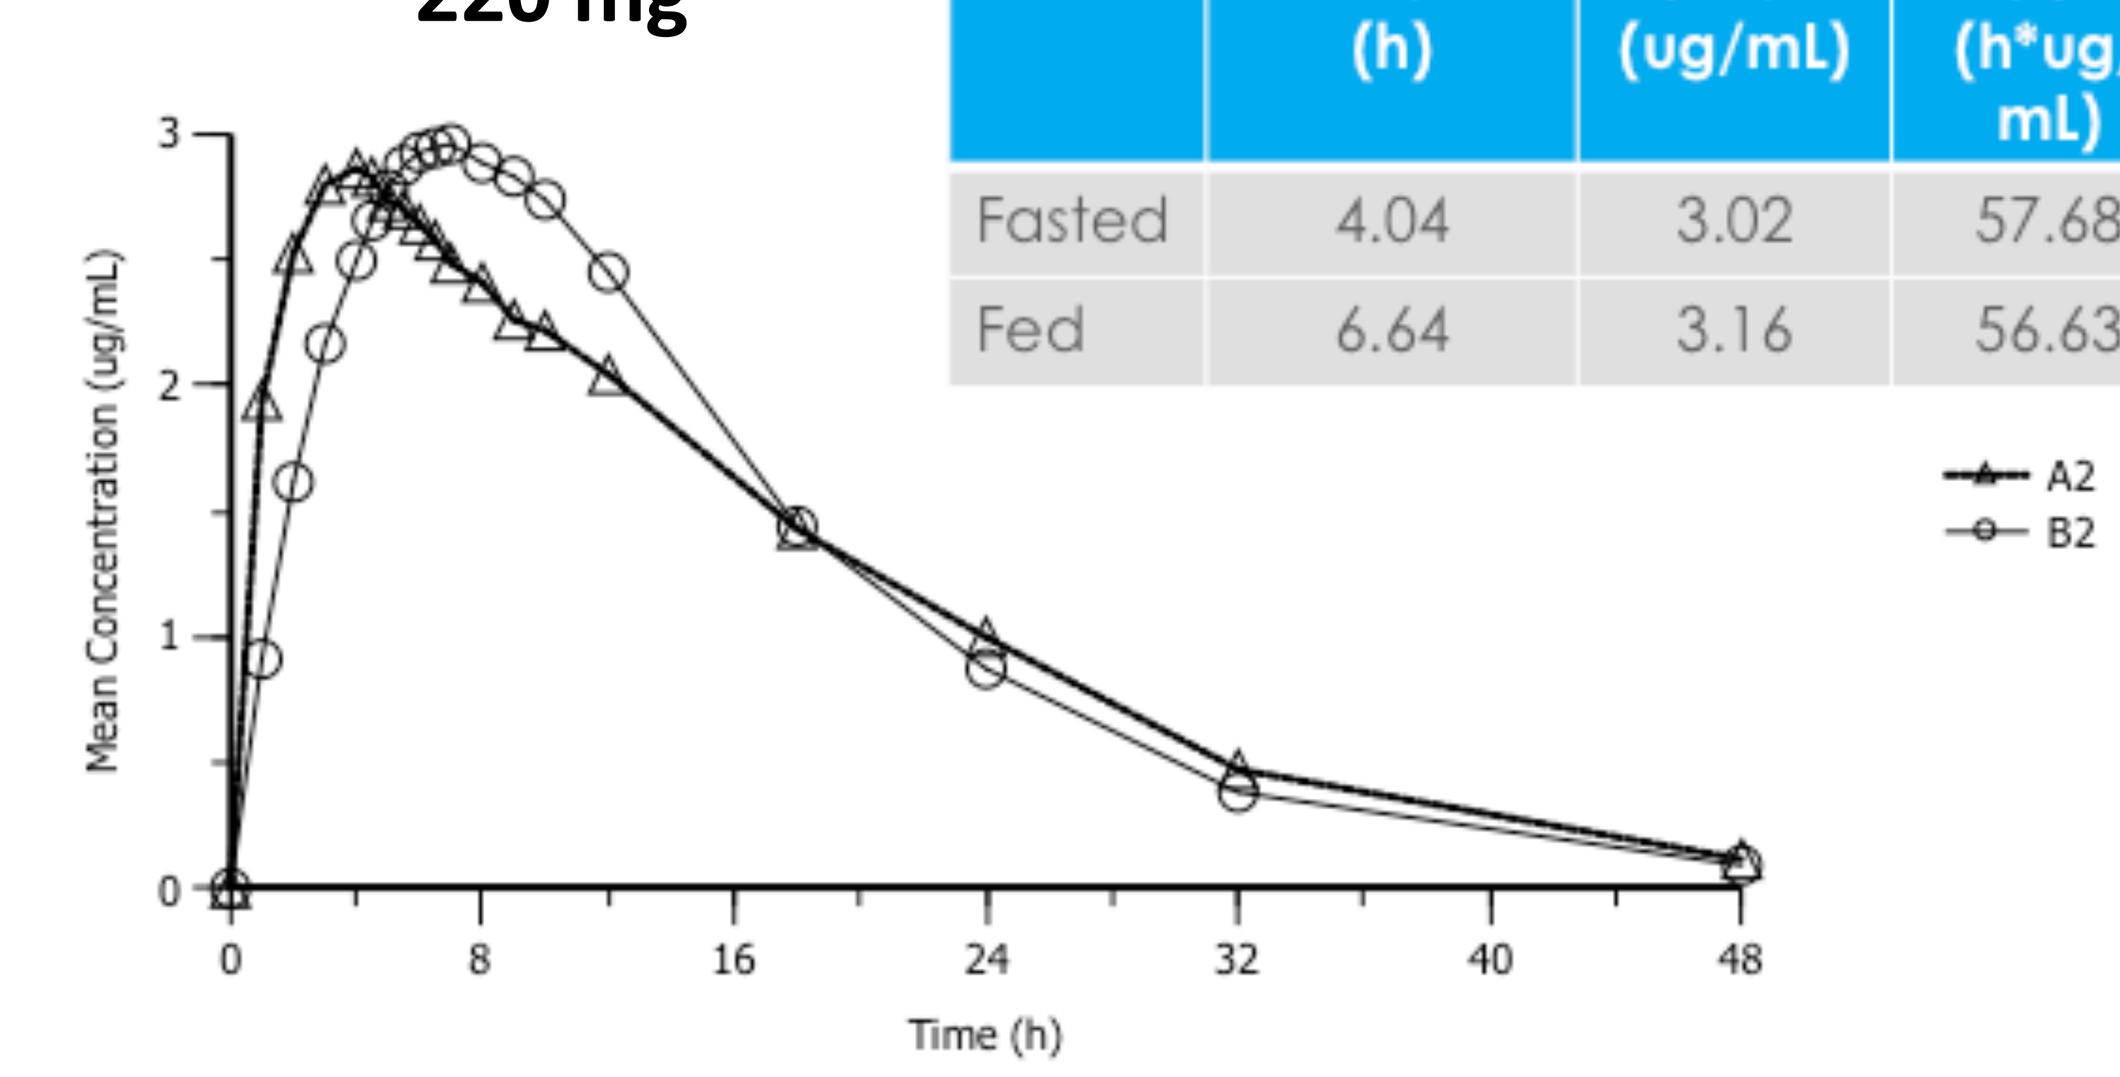
Mean Concentration (ug/mL)
Time (h)
A2
B2

	Tmax (h)	Cmax (ug/mL)	AUCinf (h*ug/mL)	T1/2 (h)
Fasted	4.04	3.02	57.68	7.81
Fed	6.64	3.16	56.63	7.56

Single Doses:

- 190 mg: Cmax between 2.31 and 2.53 ug/mL (acceptable)
- 220 mg: Cmax between 3.02 and 3.16 ug/mL (preferred)

190 mg and 220 mg Extended Release AGB101 Simulated Steady State Dose PK profile (Fed)

190 mg

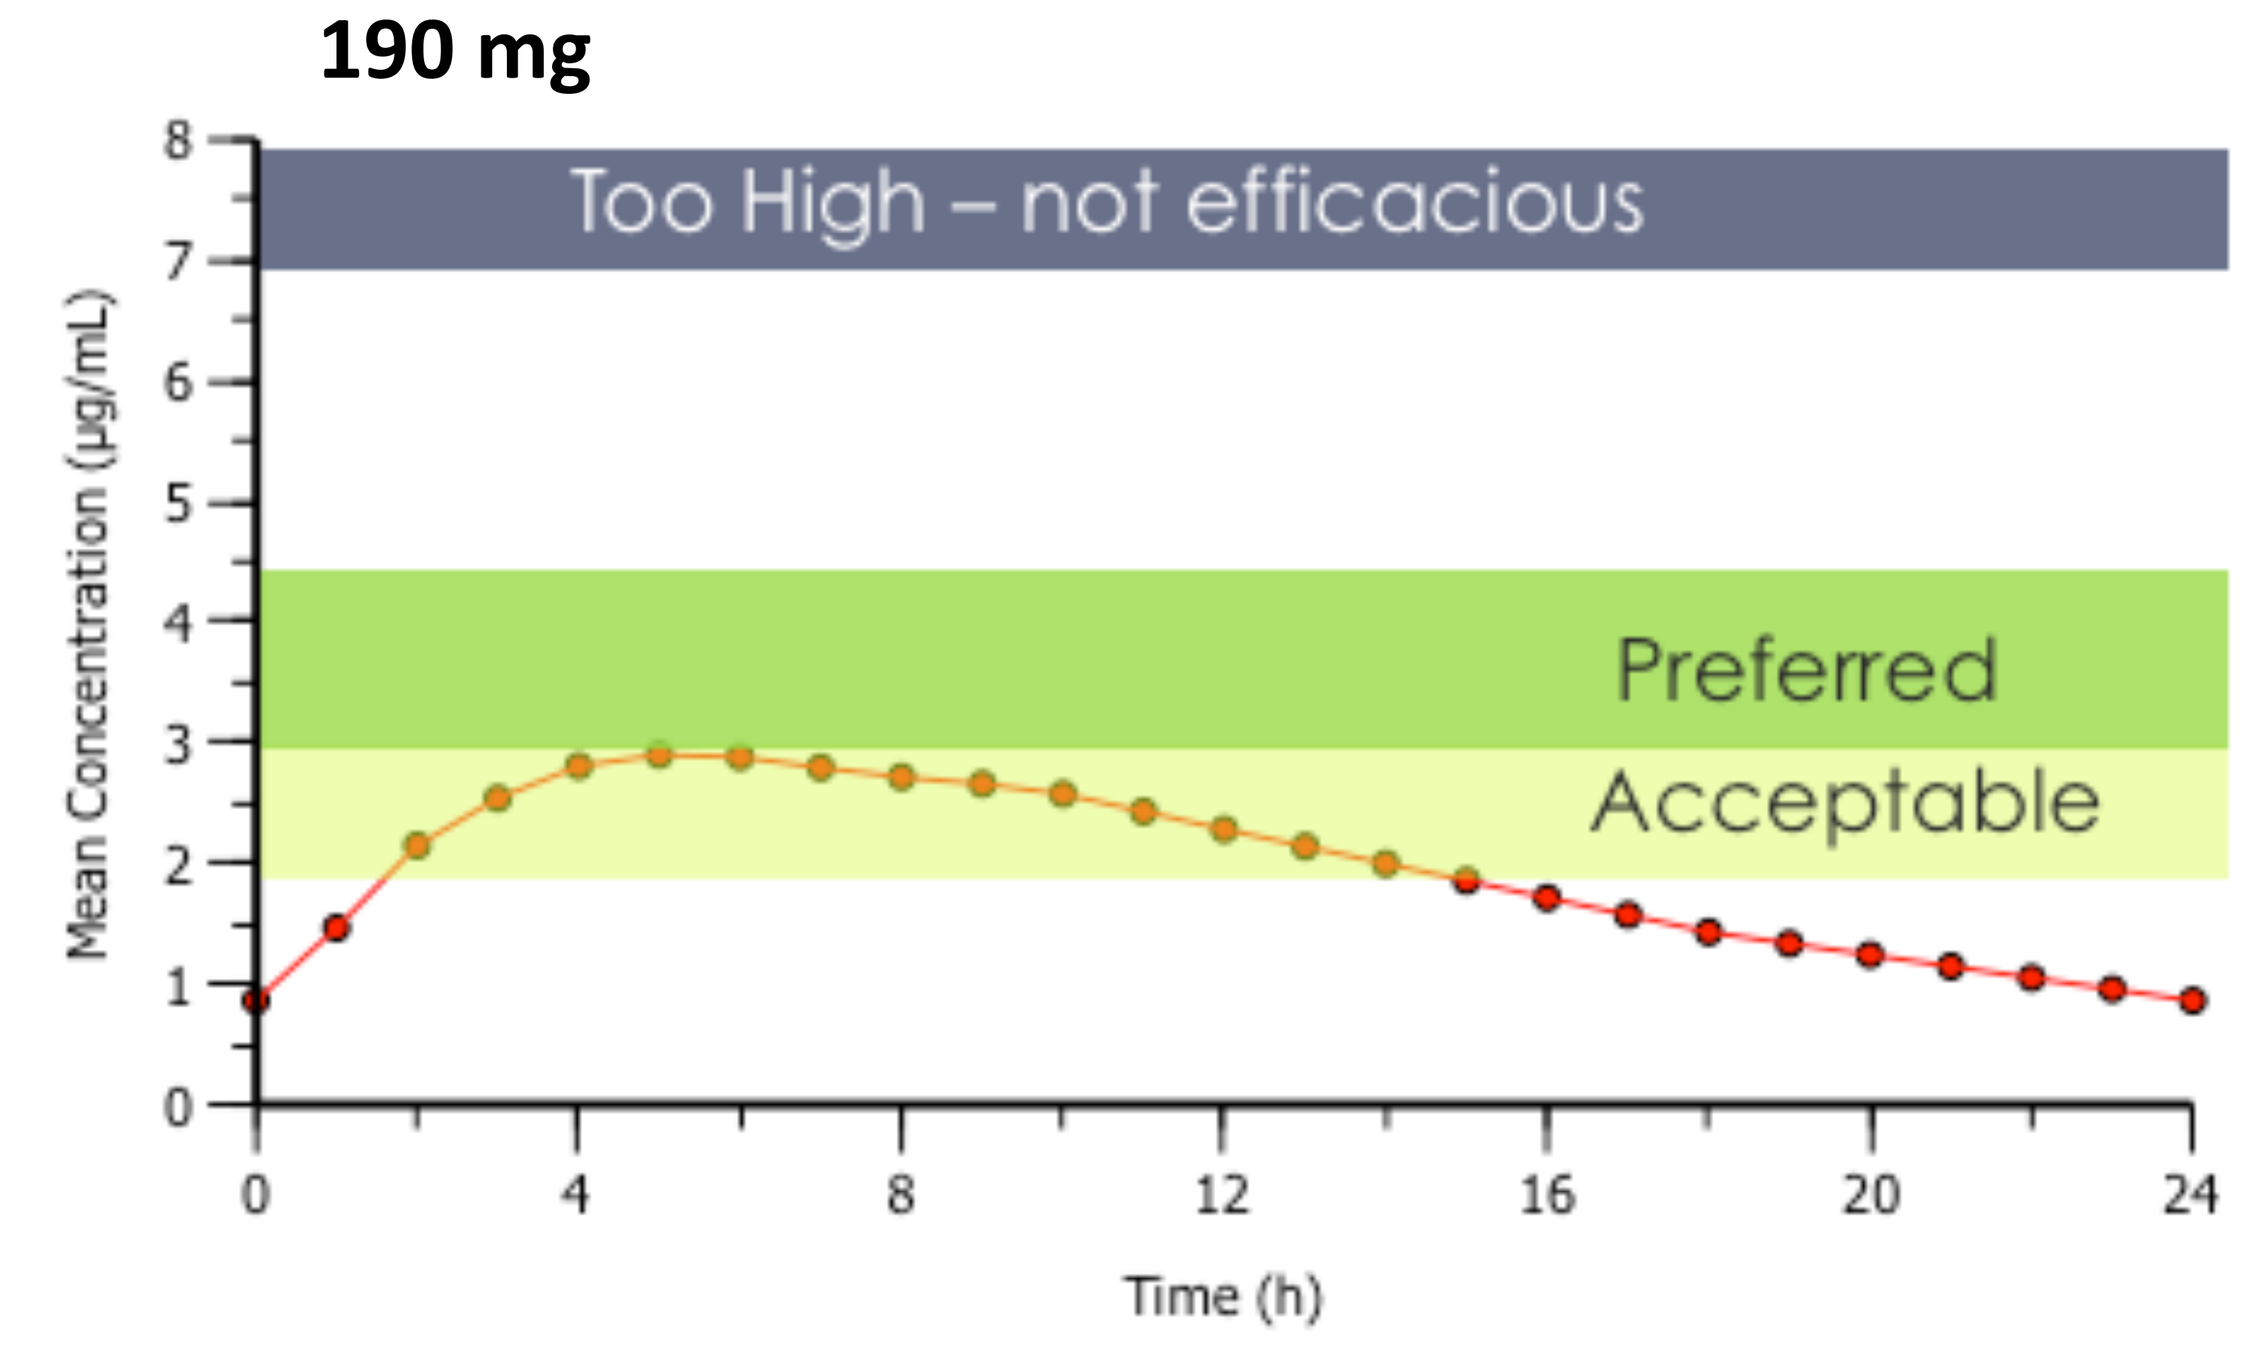
Too High – not efficacious
Preferred
Acceptable
Mean Concentration (µg/mL)
Time (h)

220 mg

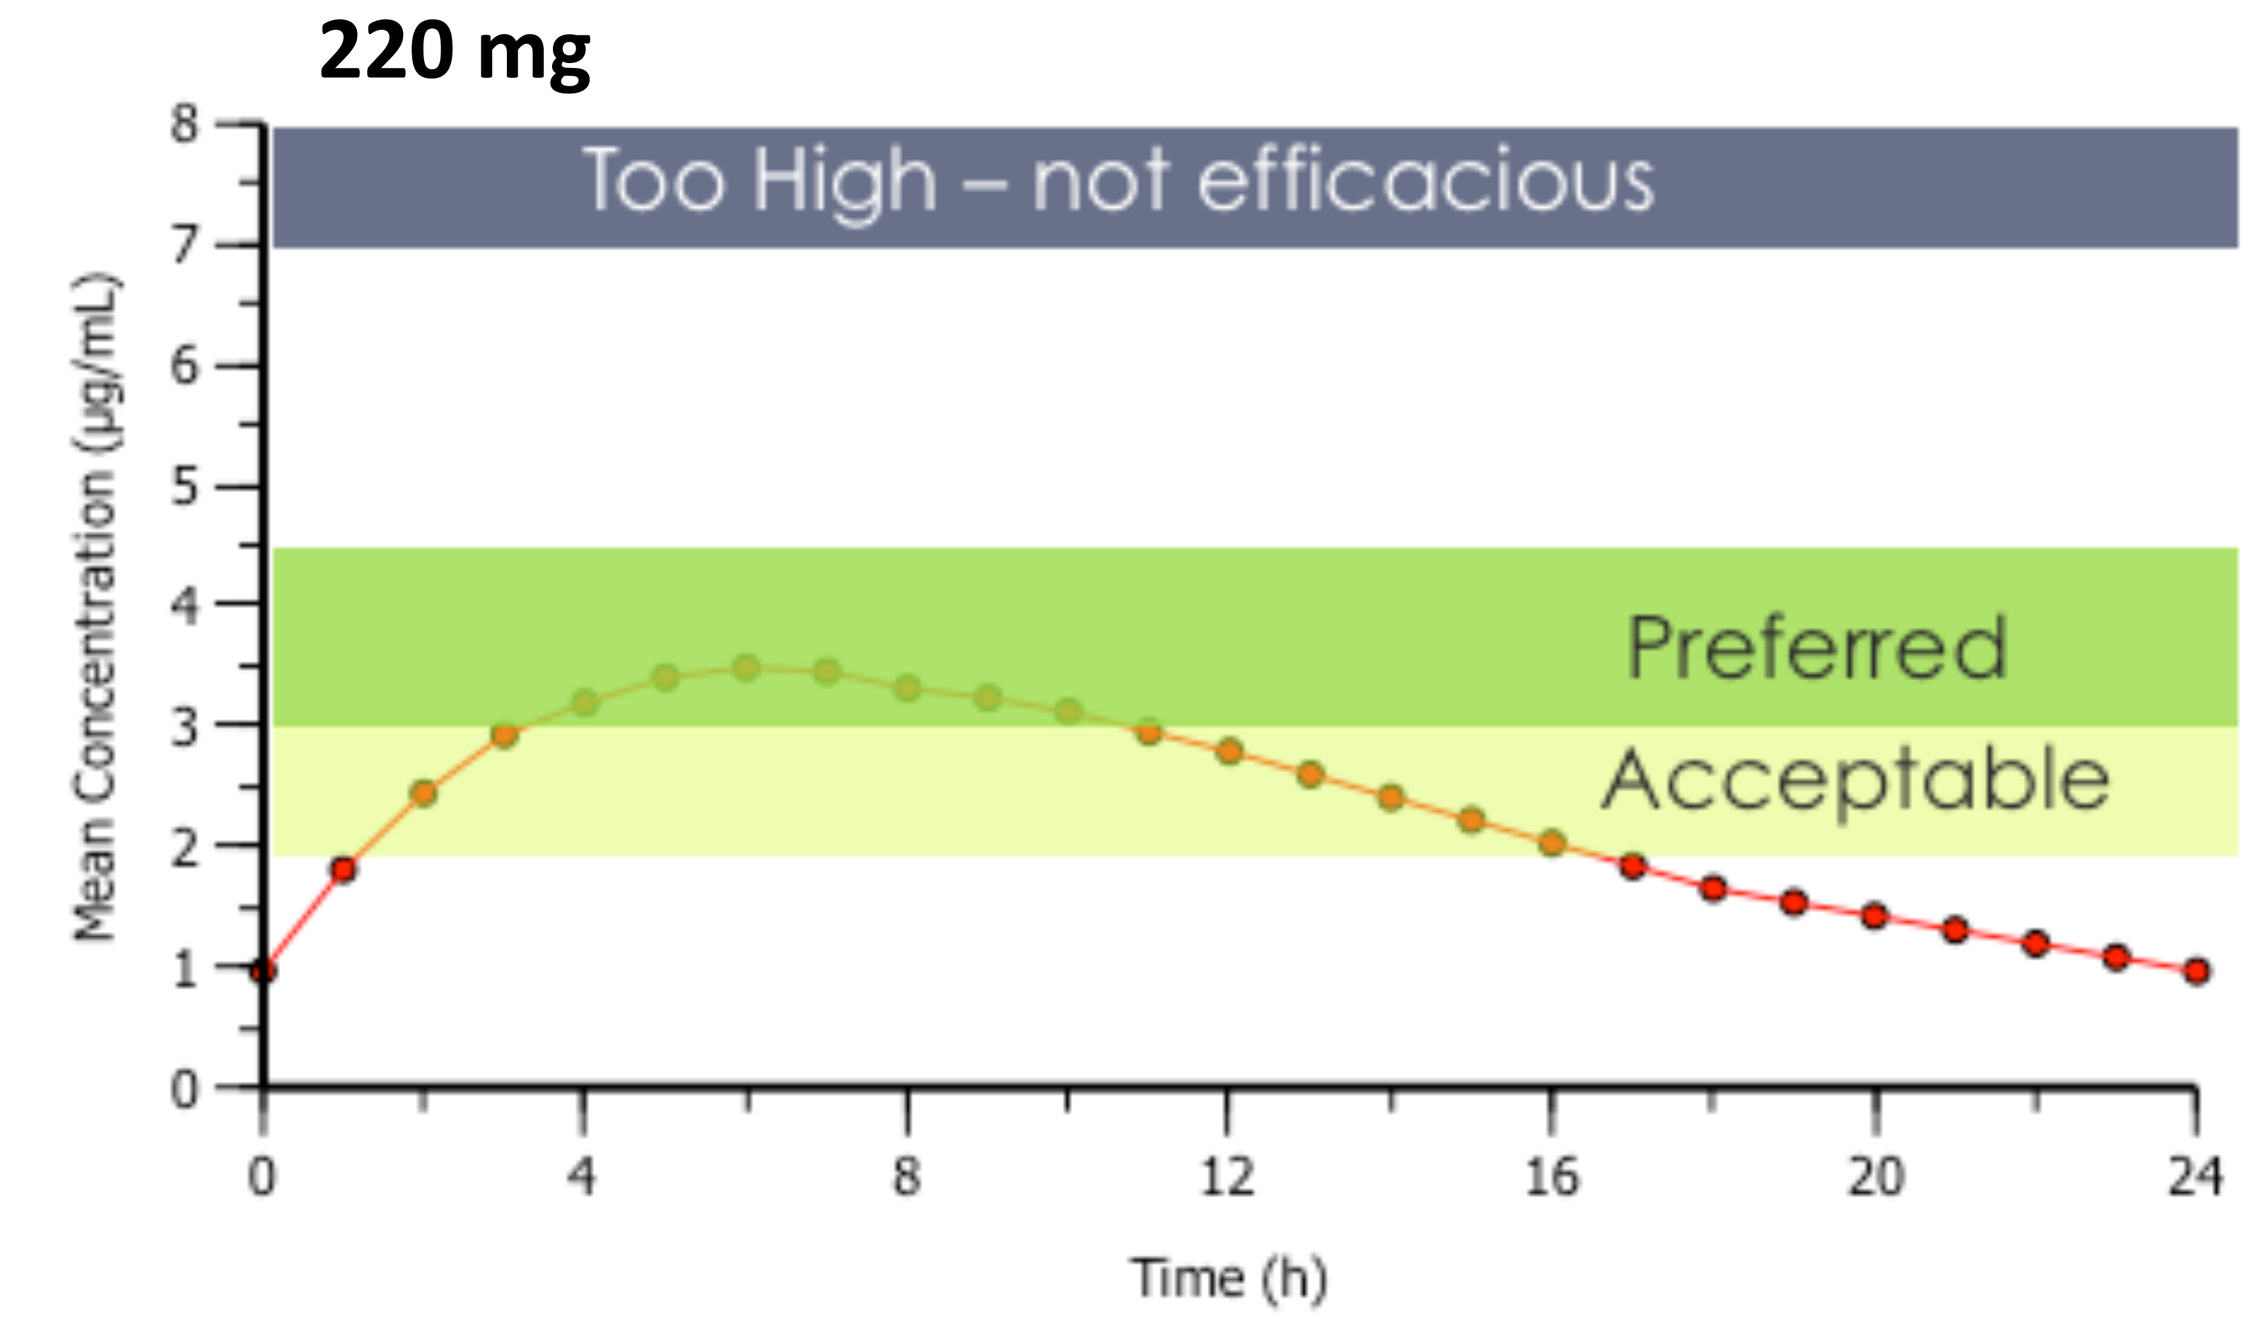
Too High – not efficacious
Preferred
Acceptable
Mean Concentration (µg/mL)
Time (h)

220 mg formulation meets Preferred Range Goals

- Exposures maintained between 2.9 and 4.4 ug/mL for 7-8 hours; above 1.9 for 14 hours.

190 mg formulation meets Acceptable Range Goals

- Exposures maintained between 1.9 and 2.9 ug/mL

Summary:

Food-Effect

The 90% confidence intervals for the log-transformed exposure parameters C_{max}, AUC_{last}, and AUC_{inf} were within the 80% to 125% range for the 190 mg and 220 mg doses. The presence of food did not alter the pharmacokinetics of the 190 mg and 220 mg levetiracetam doses. Cmax was delayed by ~ 2.5 hours.

Dose Proportionality

The 90% confidence intervals for the dose-normalized log-transformed exposure parameters C_{max}/D, AUC_{last}/D, and AUC_{inf}/D were within the 80% to 125% range for fed and fasted conditions. Levetiracetam exposure, as measured by C_{max}/D, AUC_{last}/D, and AUC_{inf}/D, increased proportionately from 190 mg to 220 mg AGB101.

Exposure Goals

220 mg met Preferred Range Goals (Exposures between 2.9 and 4.4 ug/mL. 190 mg met Acceptable Range Goals (Exposures between 1.9 and 2.9 ug/mL.

Conclusions:

AGB101 (220 mg) met all preset criteria. Sustained plasma levels of levetiracetam consistent with Phase II efficacy were observed over a 14-hour period. The PK profile was consistent with an extended release formulation suitable for once daily dosing. There were no food effects on Cmax or AUC. This dose/formulation is suitable for use in our upcoming Phase III program in aMCI.

Funded by:
Alzheimer's Drug Discovery Foundation